Generation of anti-LAG-3 monoclonal antibodies for use in immunotherapy combinations

H. Toni Jun[1], Patricia A. McNeeley[1], Greg Gold[1], Brandon Hynes[1], Laurence Altobell III[1], Mark Chhoa[1], Marilyn R. Kehry[1], Yan Wang[2], Haley Laken[2] and David J. King[1]
[1]AnaptysBio, San Diego CA and [2]TESARO Waltham, MA

Abstract

Inhibitory immune checkpoints maintain self-tolerance in the normal immune system but can be co-opted in cancer to allow tumor escape from immune surveillance. Validation of immune checkpoint inhibitors has been provided by antibodies (mAbs) that inhibit the CTLA-4 and PD-1 pathways, which have shown significant clinical activity alone and in combination. In mouse models, blockade of other T-cell inhibitory signaling checkpoints such as LAG-3 has also been shown to be effective. Combination anti-PD-1 and anti-LAG-3 therapy was explored *in vivo* in MC38 and Colon26 mouse syngeneic tumor models with mAbs to mouse PD-1 and LAG-3. In the MC38 model, the tumor-free animals were increased from 7/10 in the anti-PD-1 alone arm to 10/10 in the combination. In the Colon26 model, the numbers of tumor-free animals in the anti-LAG-3/anti-PD-1 combination group were 10/12 while anti-PD-1 alone had 3/12 tumor-free animals.
mAbs that inhibit human LAG-3 signaling were identified by screening the AnaptysBio Evolvable Library of fully human mAbs using a soluble LAG-3. We also generated humanized mouse mAbs to LAG-3. The resulting mAbs were matured to high affinity and potency via SHM-XEL™ which uses mammalian cell display of human IgG followed by *in vitro* somatic hypermutation. In *in vitro* cell assays, inhibition of LAG-3 alone demonstrates immune stimulatory activity as evidenced by increased IL-2 secretion in a mixed lymphocyte reaction (MLR). Combination of an anti-LAG-3 antibody with an anti-PD-1 antibody could shift the EC_{50} approximately 17-fold depending upon donor.
Overall, these data support combination cancer immunotherapy with anti-LAG-3 and anti-PD-1.

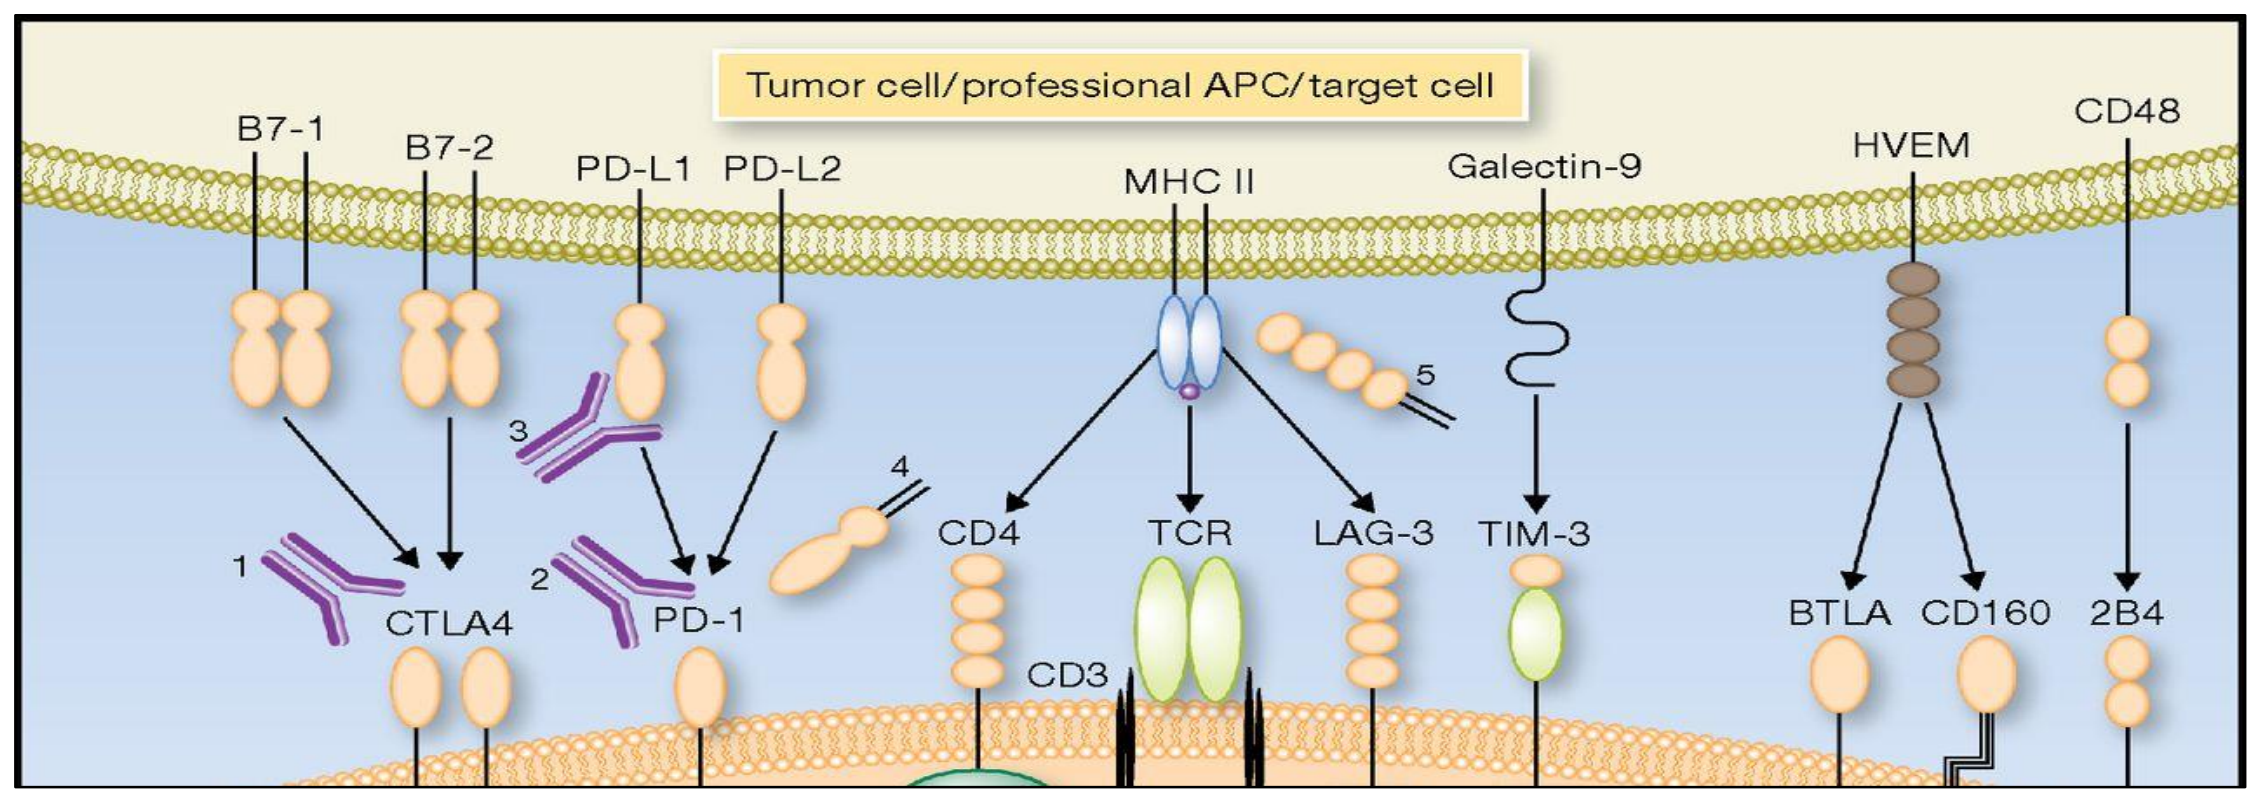
Tumor cell/professional APC/target cell
B7-1
B7-2
PD-L1
PD-L2
MHC II
Galectin-9
HVEM
CD48
CTLA4
PD-1
CD4
TCR
LAG-3
TIM-3
CD3
BTLA
CD160
2B4

Overview of select immune checkpoint molecules.
PD-1 ligation by PD-L1 or PD-L2 results in membrane proximal decreases in TCR signaling. LAG-3 signaling is dependent on interaction with its ligand, MHC II to inhibit T-cell signaling.
Nirschl and Drake (2013).

PD-1 and LAG-3 are Co-Expressed on Activated T-cells

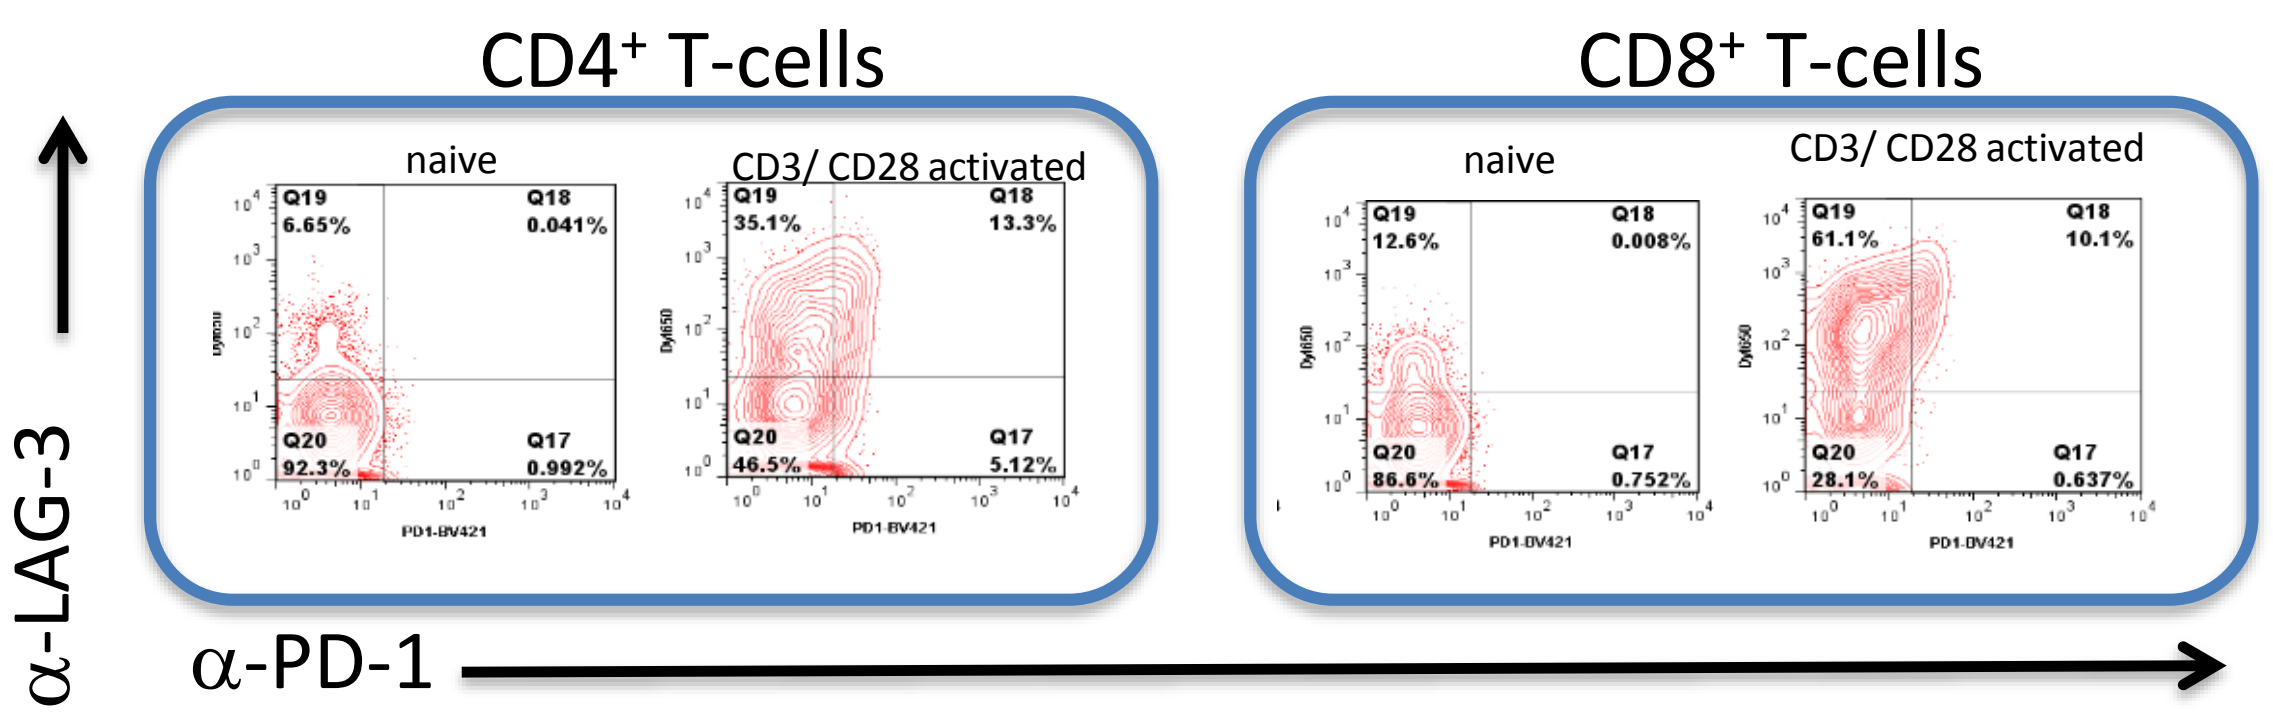
CD4+ T-cells
CD8+ T-cells
naive
CD3/ CD28 activated
α-LAG-3
α-PD-1

Human donor CD4+ or CD8+ T-cells were isolated and activated using plate-bound anti-CD3 and anti-CD28 mAbs. After 48 hours, cells were stained for PD-1 and LAG-3. Increased levels of PD-1/LAG-3 double positive cells were observed after stimulation compared to naïve T-cells.

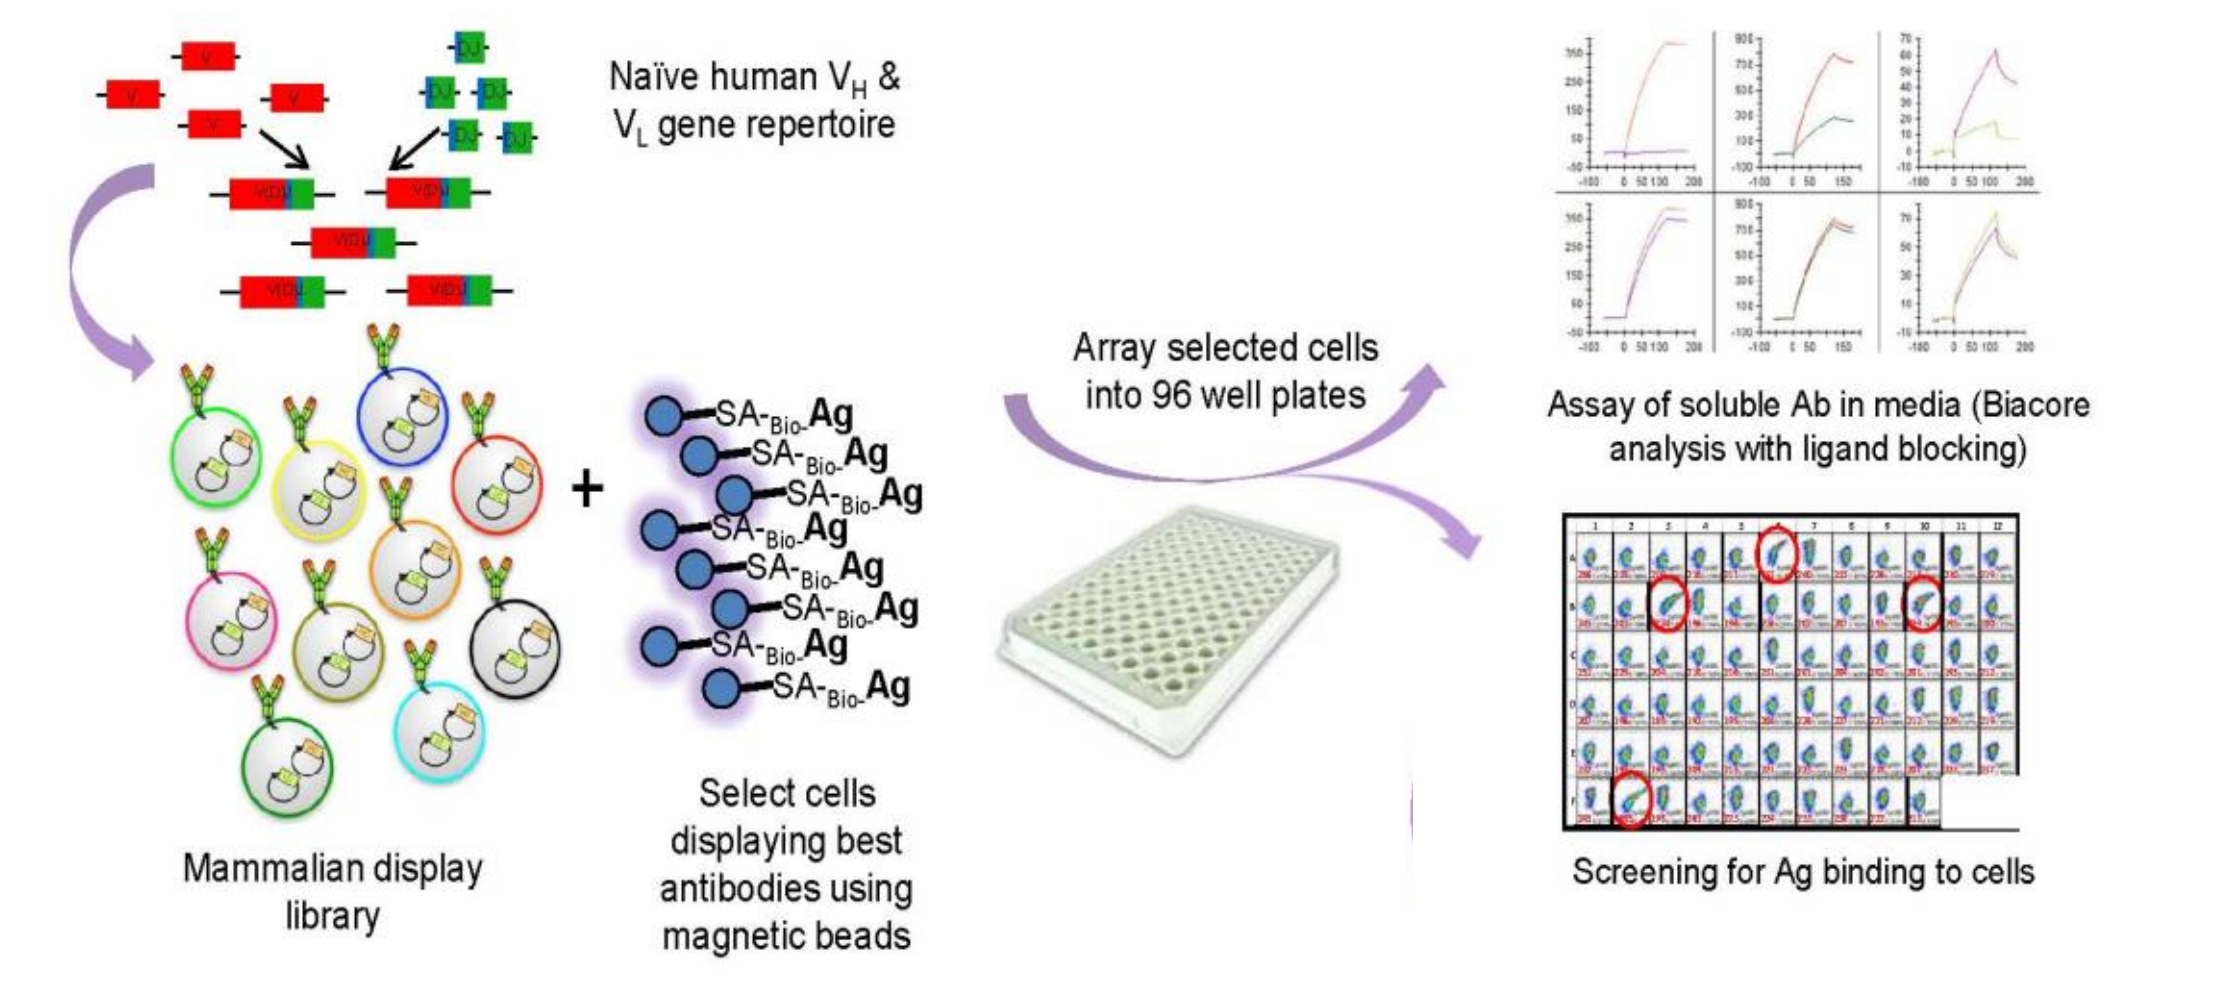
Naïve human V_H & V_L gene repertoire
Mammalian display library
Select cells displaying best antibodies using magnetic beads
Array selected cells into 96 well plates
Assay of soluble Ab in media (Biacore analysis with ligand blocking)
Screening for Ag binding to cells

ABEL library screen overview.
Cells expressing surface-displayed, fully-human antibodies (Abs) that bind to soluble human LAG-3 ECD were identified from a screening campaign of the ABEL library using magnetic beads coated with huLAG-3. A panel of fully-human germline antibodies were isolated that bound specifically to LAG-3. The heavy (HC) and light (LC) chains of select hits were retransfected to create a stable cell line displaying the mAb on the cell surface. Activation induced cytodine deaminase (AID) was then transfected to begin somatic hypermutation (SHM). Iterative rounds of FACS sorting with decreasing concentrations of antigen were performed to identify mAbs with increased affinity for LAG-3.

Complete Tumor Regressions with anti-LAG3 and anti-PD1 Combination in the MC38 Syngeneic Tumor Model

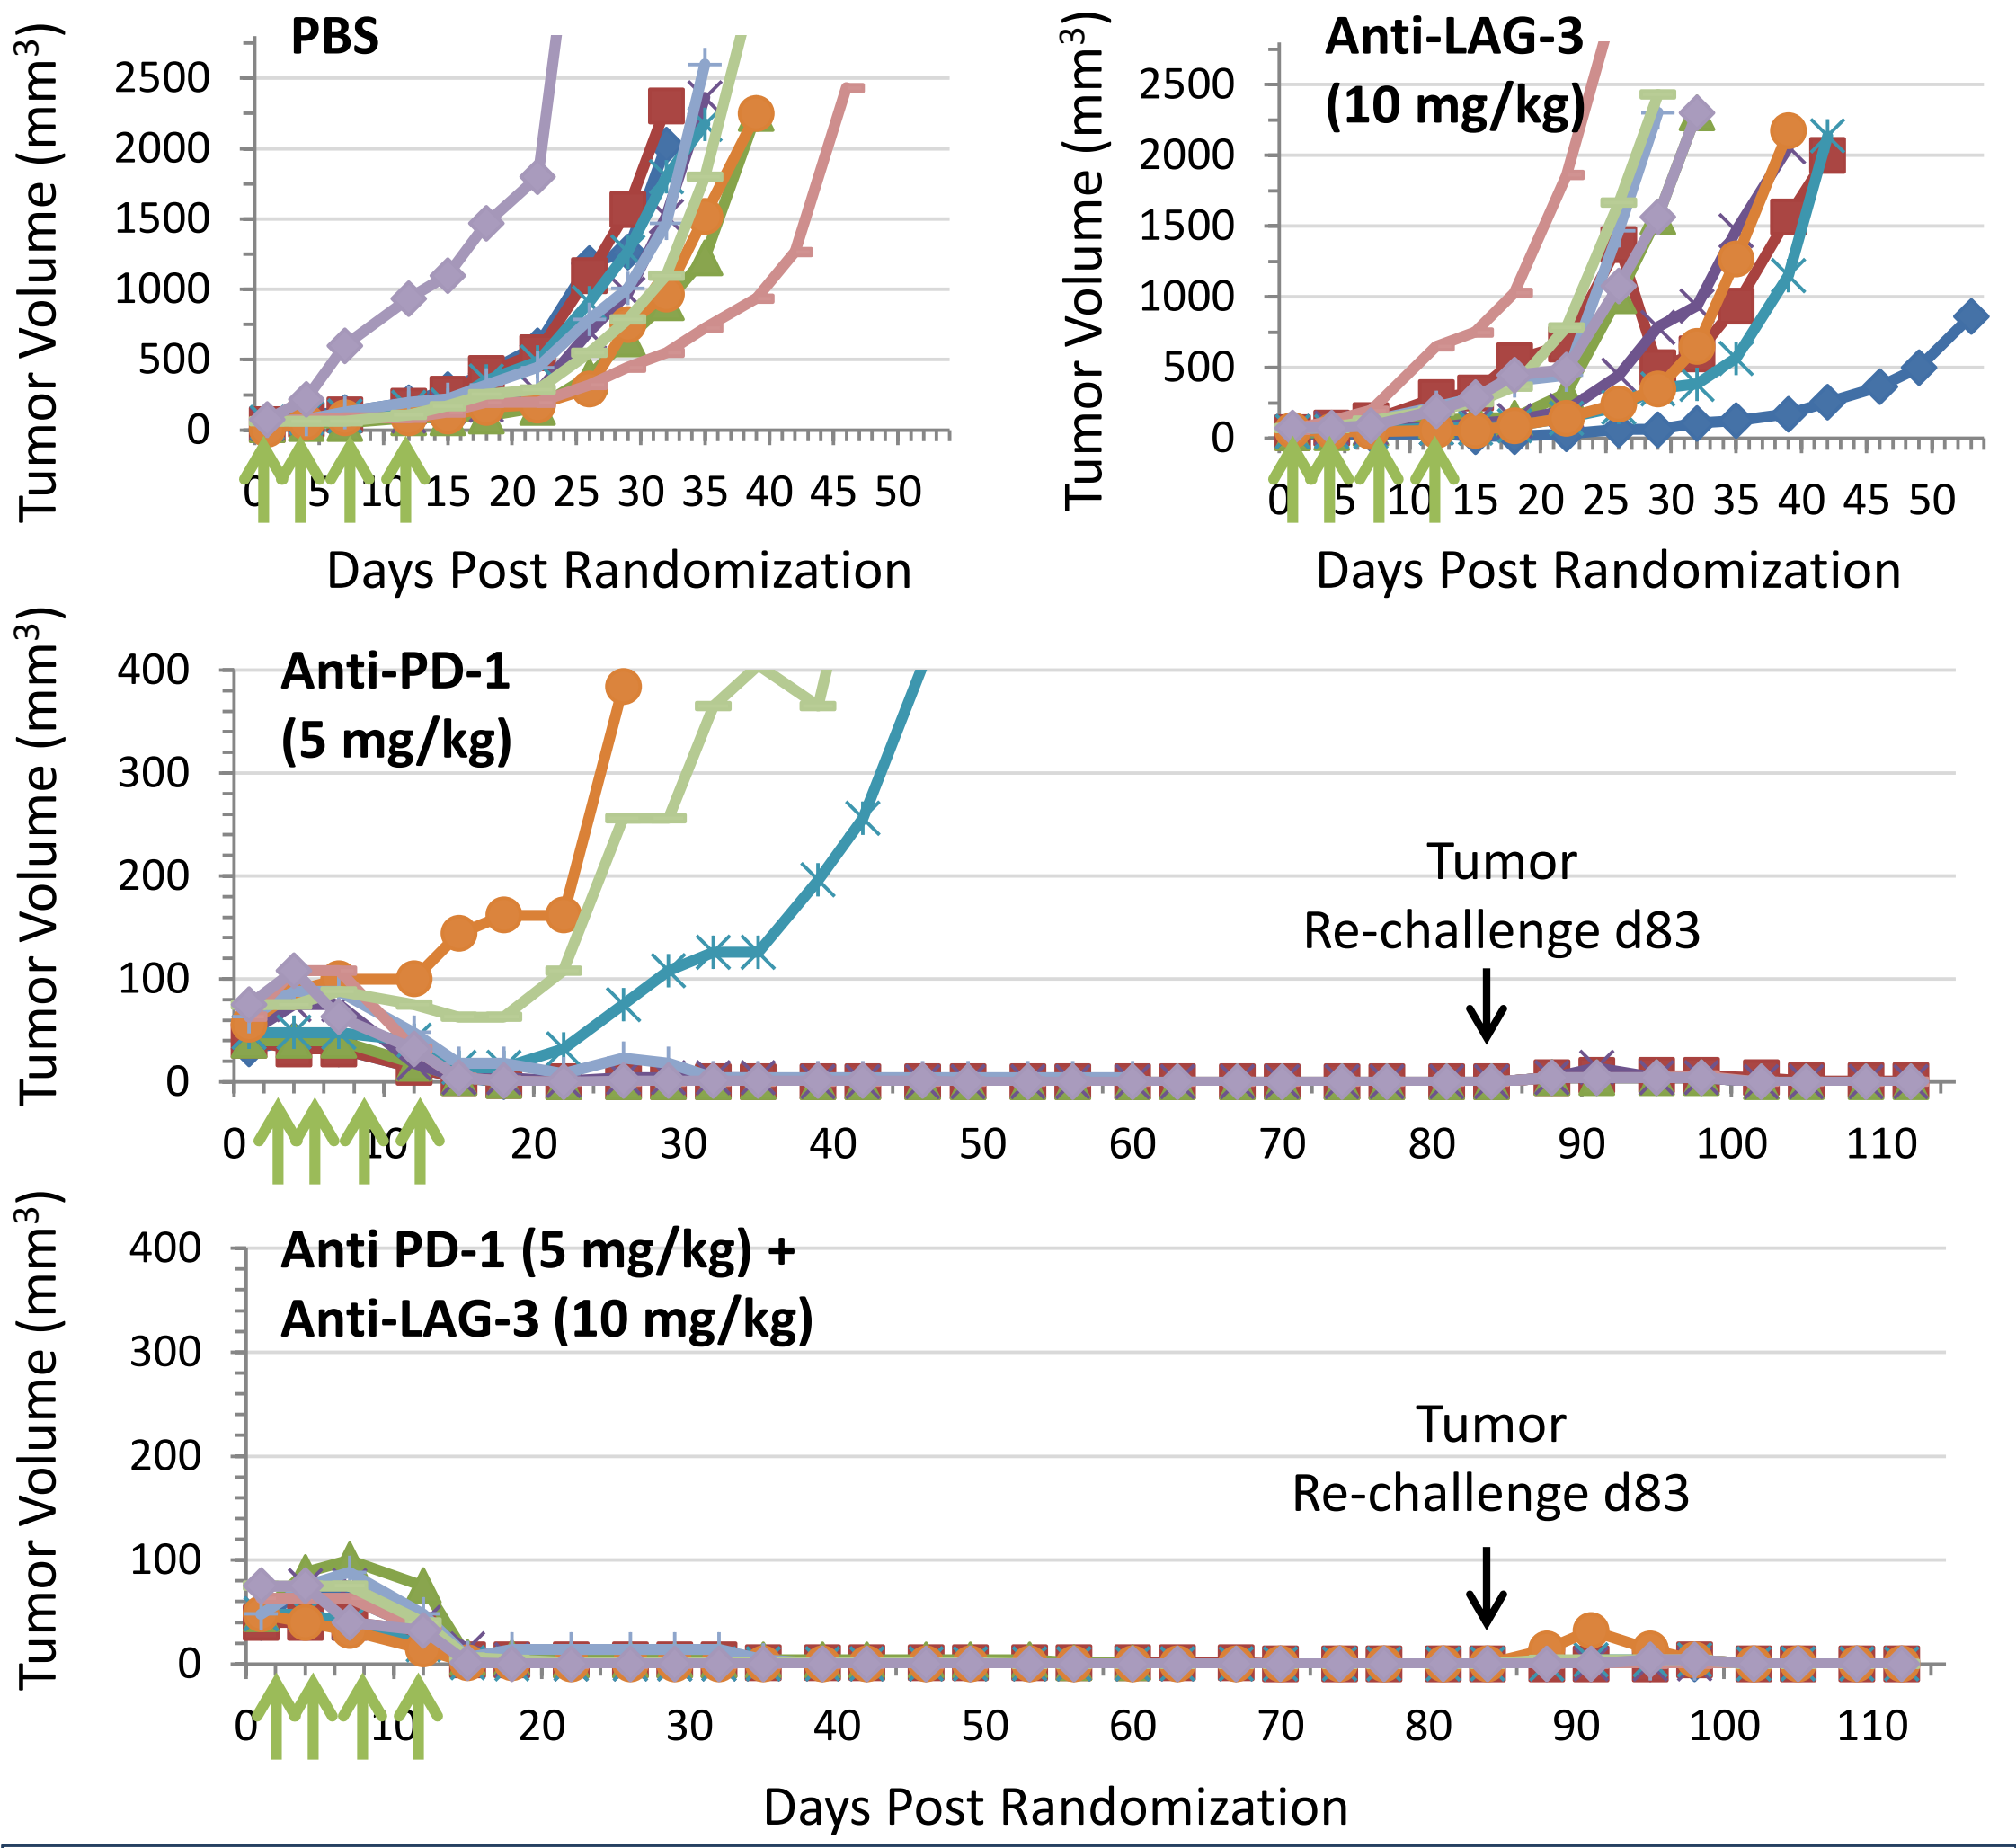
PBS
Anti-LAG-3 (10 mg/kg)
Anti-PD-1 (5 mg/kg)
Anti PD-1 (5 mg/kg) + Anti-LAG-3 (10 mg/kg)
Tumor Re-challenge d83
Tumor Volume (mm3)
Days Post Randomization

10 animals in each group were implanted with MC38 cells and dosed on d1, d4, d7, and d11 post randomization for size. All 10 animals in the combination group were tumor free compared with 7 animals in the anti-PD-1 (mAb RMP1-14, BioXcell) alone group and none in the anti-LAG-3 (mAb C9B7W, BioXcell) group. Tumor free animals from the anti-PD-1 and combination-treated groups were re-inoculated with 4x the number of MC38 cells. No tumors grew in either re-challenged group compared with 100% tumor take in the control group (not shown).

Complete Tumor Regressions with anti-LAG3 and anti-PD1 Combination in the Colon26 Syngeneic Tumor Model

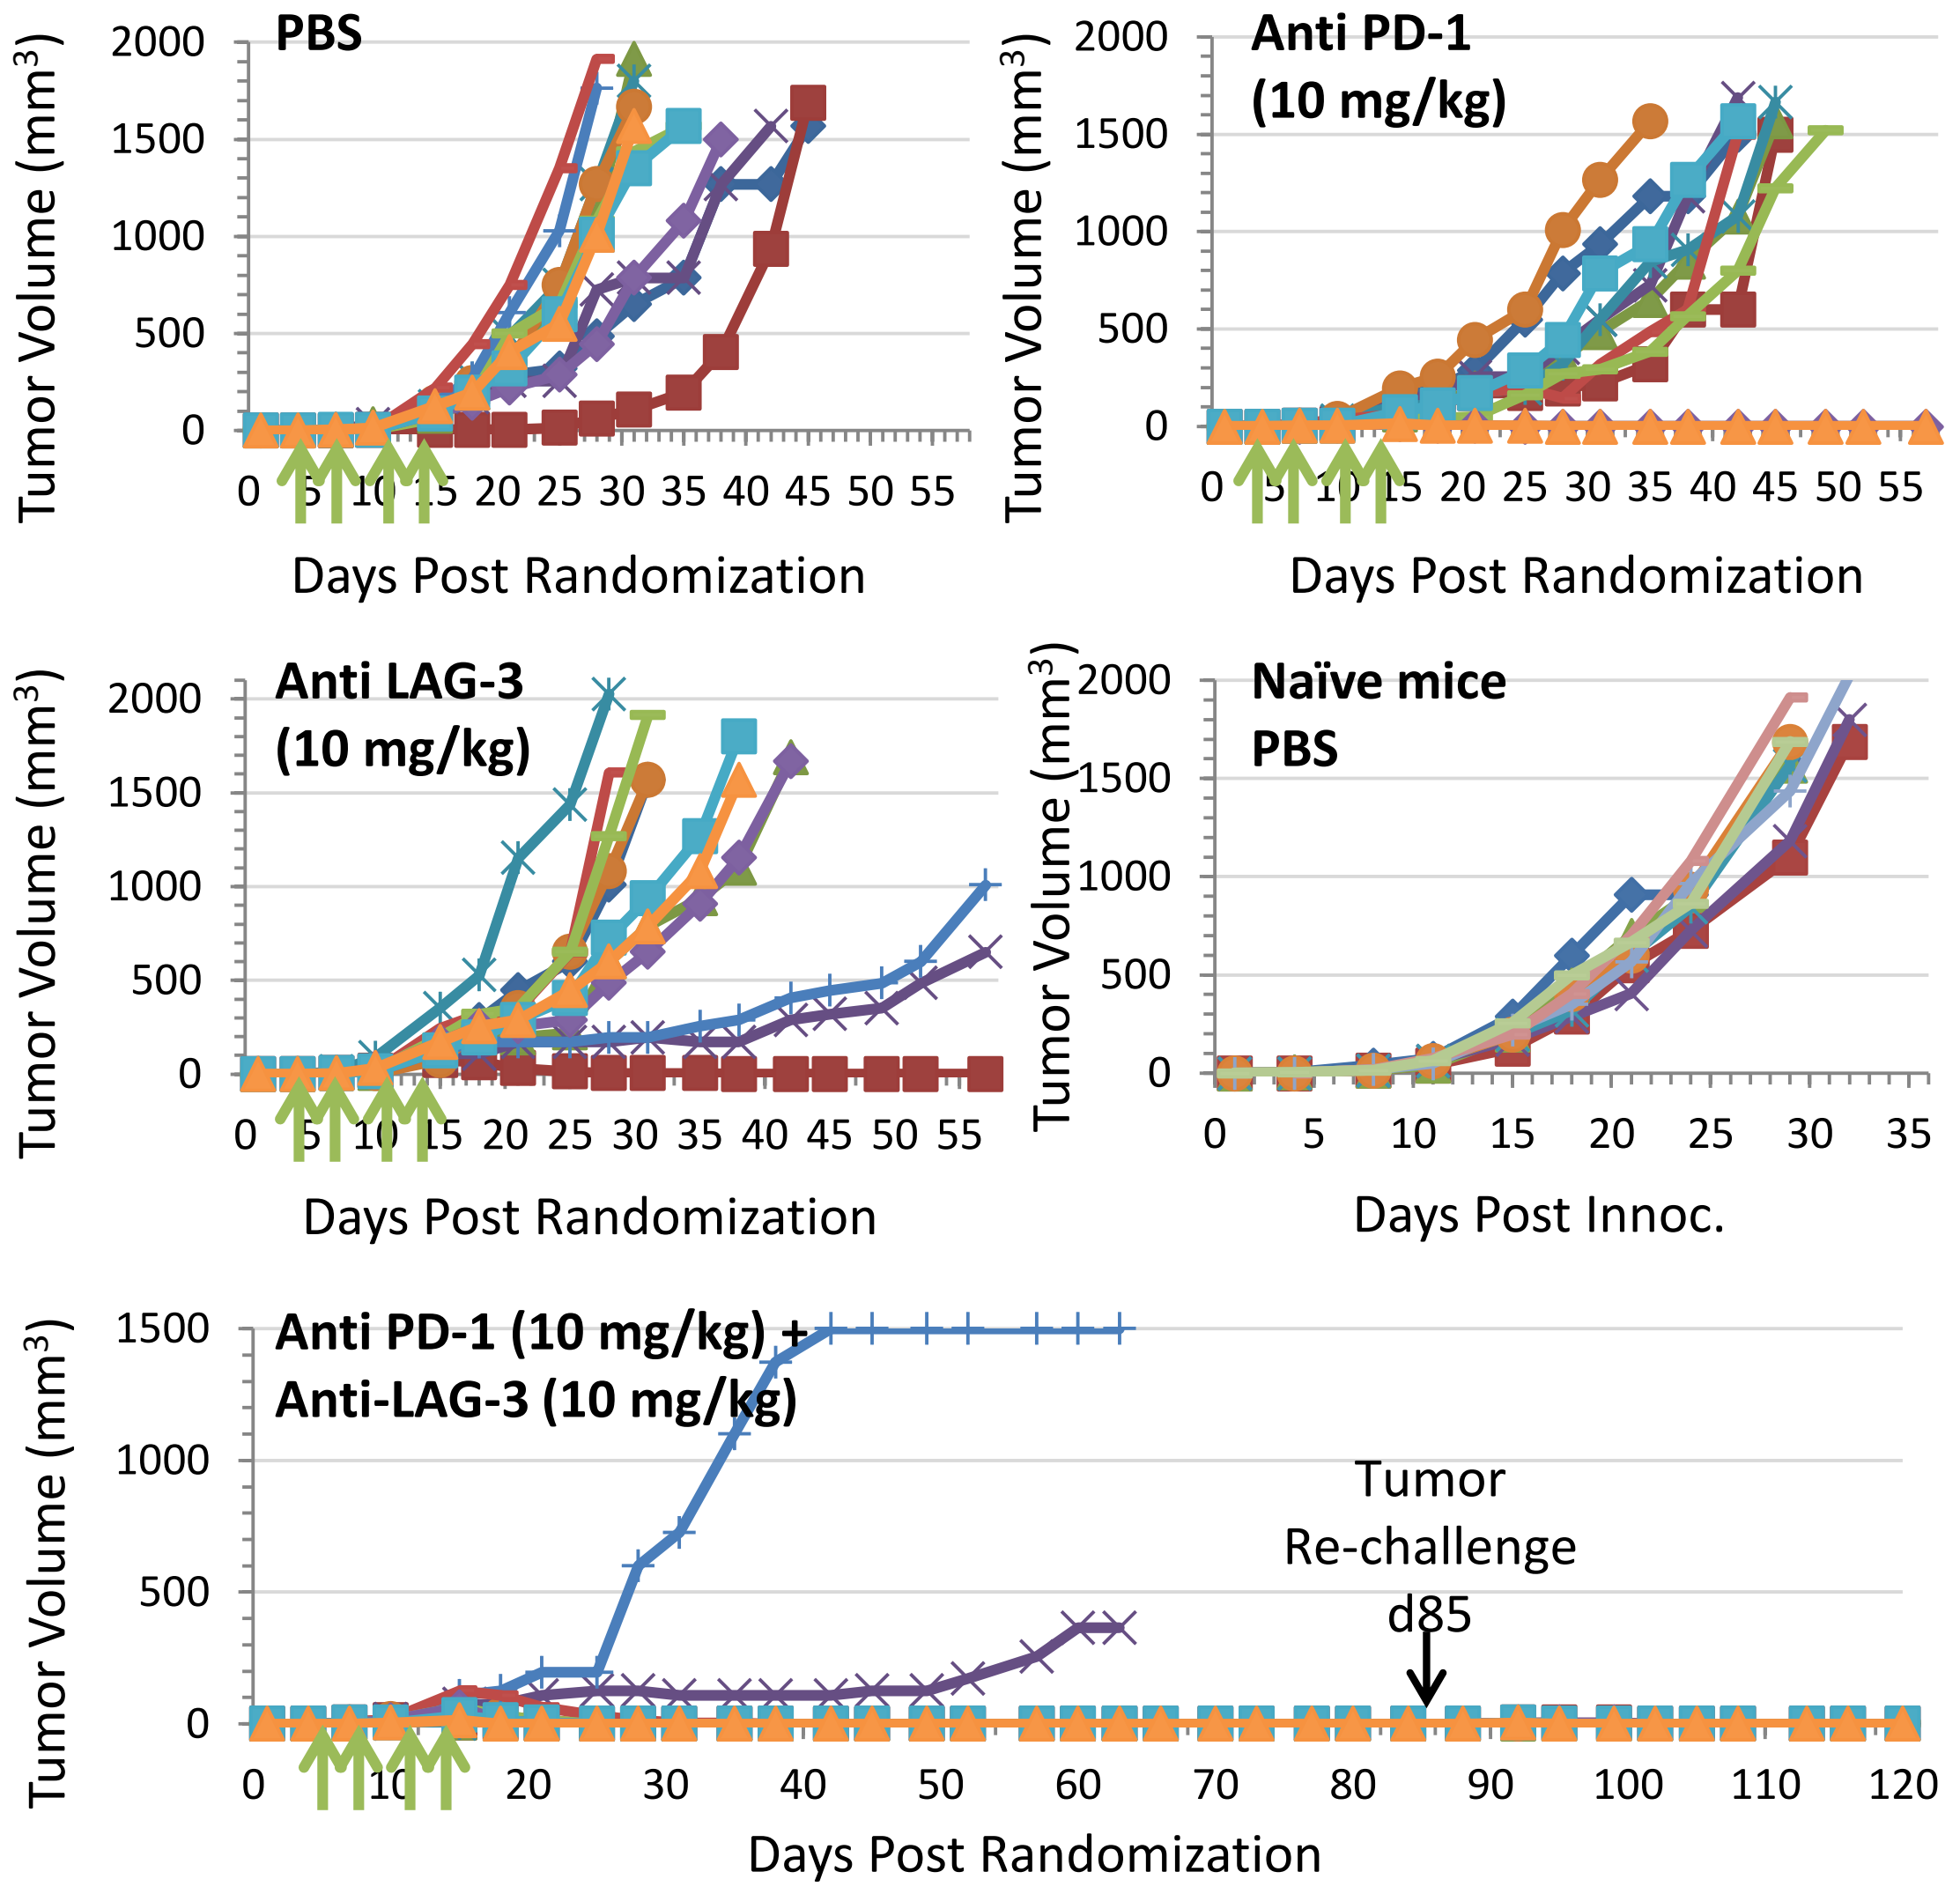
PBS
Anti PD-1 (10 mg/kg)
Anti LAG-3 (10 mg/kg)
Naïve mice PBS
Anti PD-1 (10 mg/kg) + Anti-LAG-3 (10 mg/kg)
Tumor Re-challenge d85
Tumor Volume (mm3)
Days Post Randomization
Days Post Innoc.

12 animals in each group were implanted with Colon26 cells dosed at d4, d7, d11, and d14. Complete response was observed in 10 out of 12 animals in the combination treated group compared with 3 out of 12 animals in the PD-1 alone group and one in the anti-LAG-3 group. Complete responder animals from the combination group were re-inoculated with the same number of Colon26 cells as initially implanted. No tumors grew in the re-challenged groups compared with 100% tumor take in the control group.

Increased Affinity of Anti-LAG-3 Candidate mAbs Generated by Somatic Hypermutation

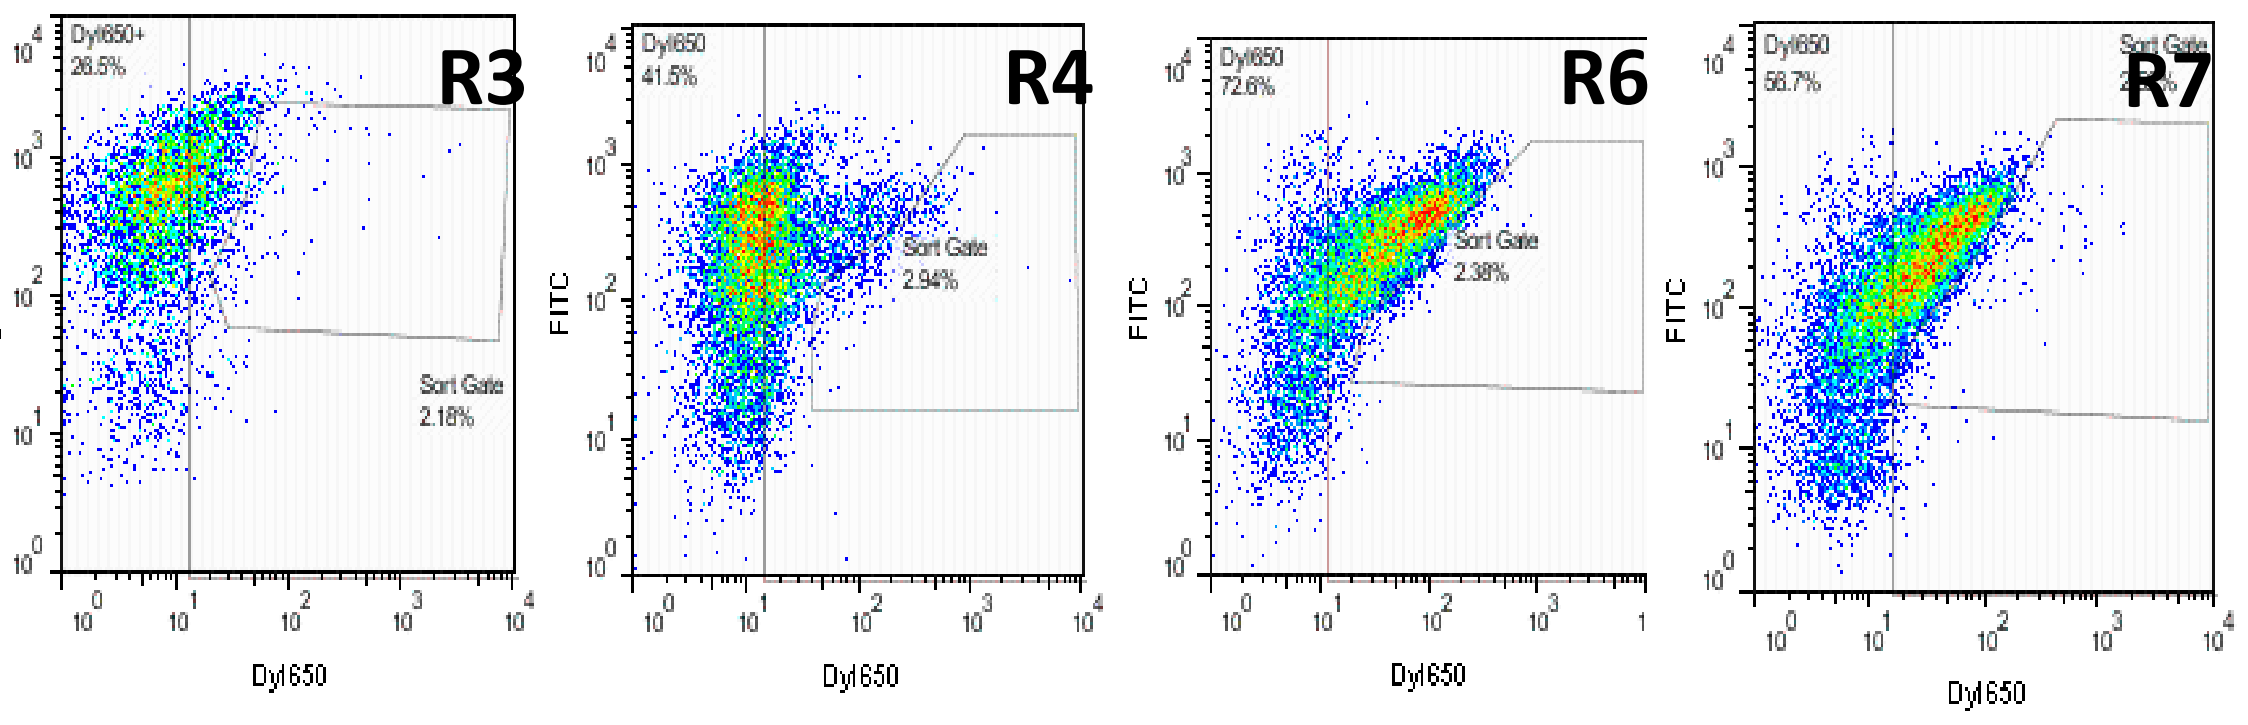
R3
R4
R6
R7
Sort Gate
Dyl650

Cells from an evolving population were sorted by flow cytometry following incubation with antigen. The gated population indicated on each FACS plot was expanded after sorting and subjected to additional rounds of cell sorting as well as sequence analysis to identify improving mAb mutations.

Lead Anti-LAG-3 mAb Inhibits the Interaction of Soluble LAG-3 with MHC Class II

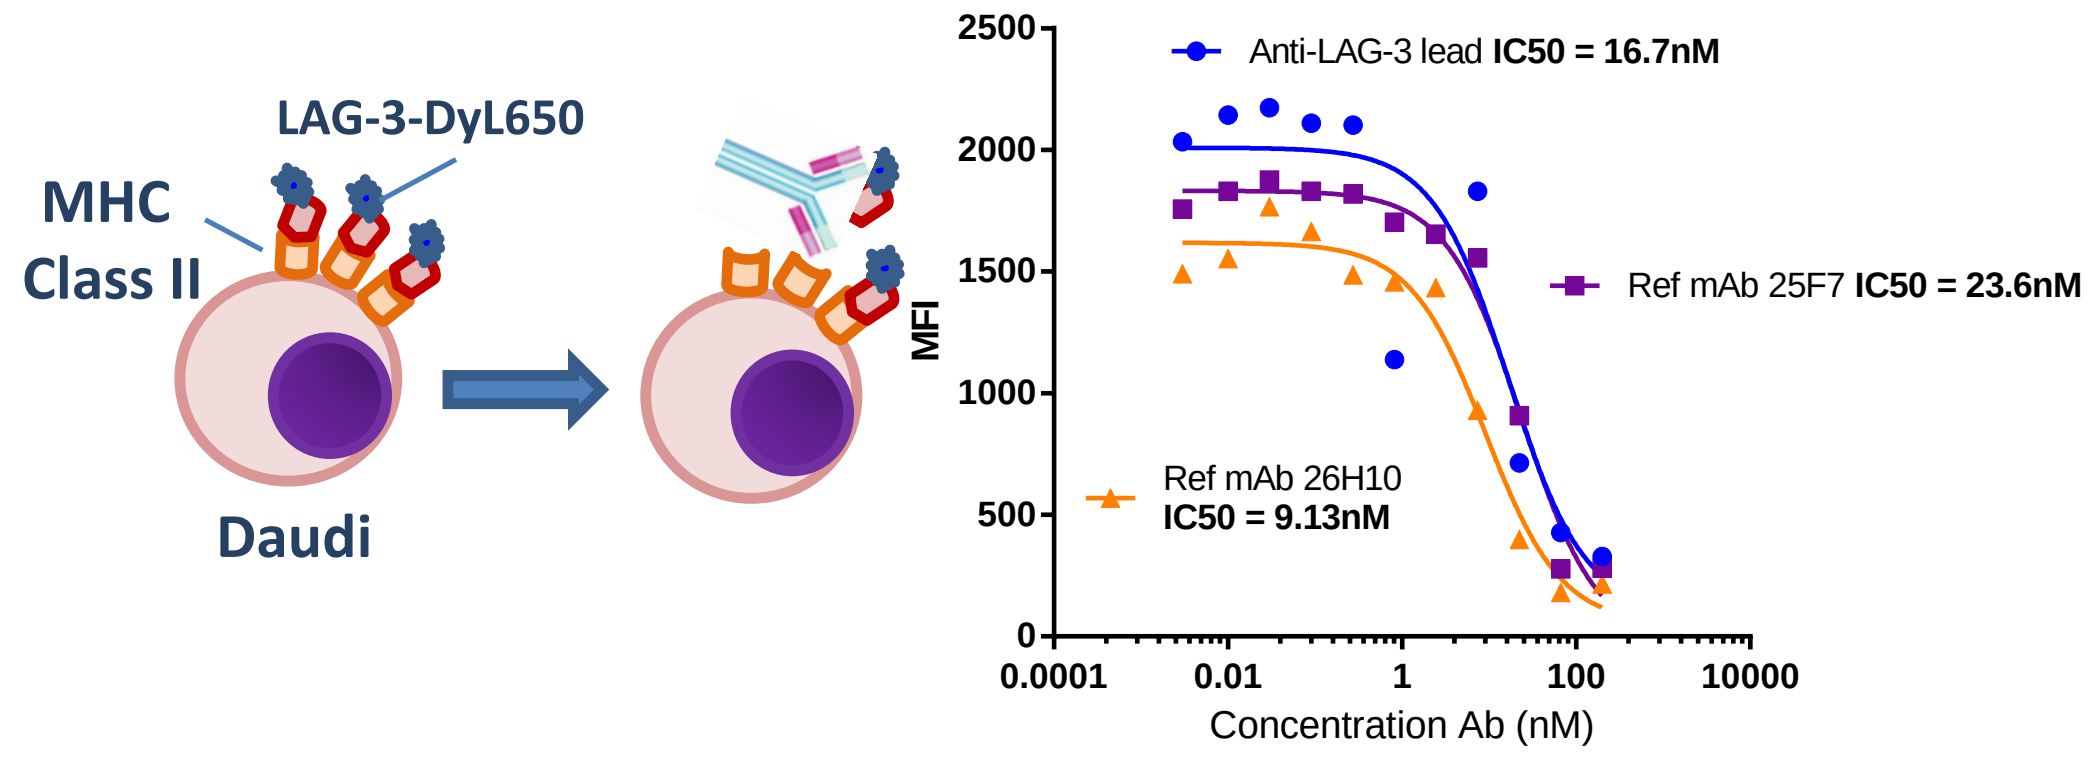
LAG-3-DyL650
MHC Class II
Daudi
Anti-LAG-3 lead IC50 = 16.7nM
Ref mAb 25F7 IC50 = 23.6nM
Ref mAb 26H10 IC50 = 9.13nM
MFI
Concentration Ab (nM)

To test the ability of an anti-LAG-3 antibody to block the LAG-3/MHC II interaction, an *in vitro* assay was developed using a soluble form of LAG-3 fused to Fc, labeled with DyL650 binding to a Daudi B-cell line, which endogenously expresses MHC II. A dose range of neutralizing antibodies was preincubated with the soluble LAG-3 and analyzed by flow cytometry. The anti-LAG-3 lead can completely inhibit the interaction of soluble LAG-3 with MHCII.

Lead Anti-LAG-3 mAb Potentiates IL-2 Secretion Alone and in Combination with an Anti-PD-1 Antagonist mAb in a Mixed Lymphocyte Reaction

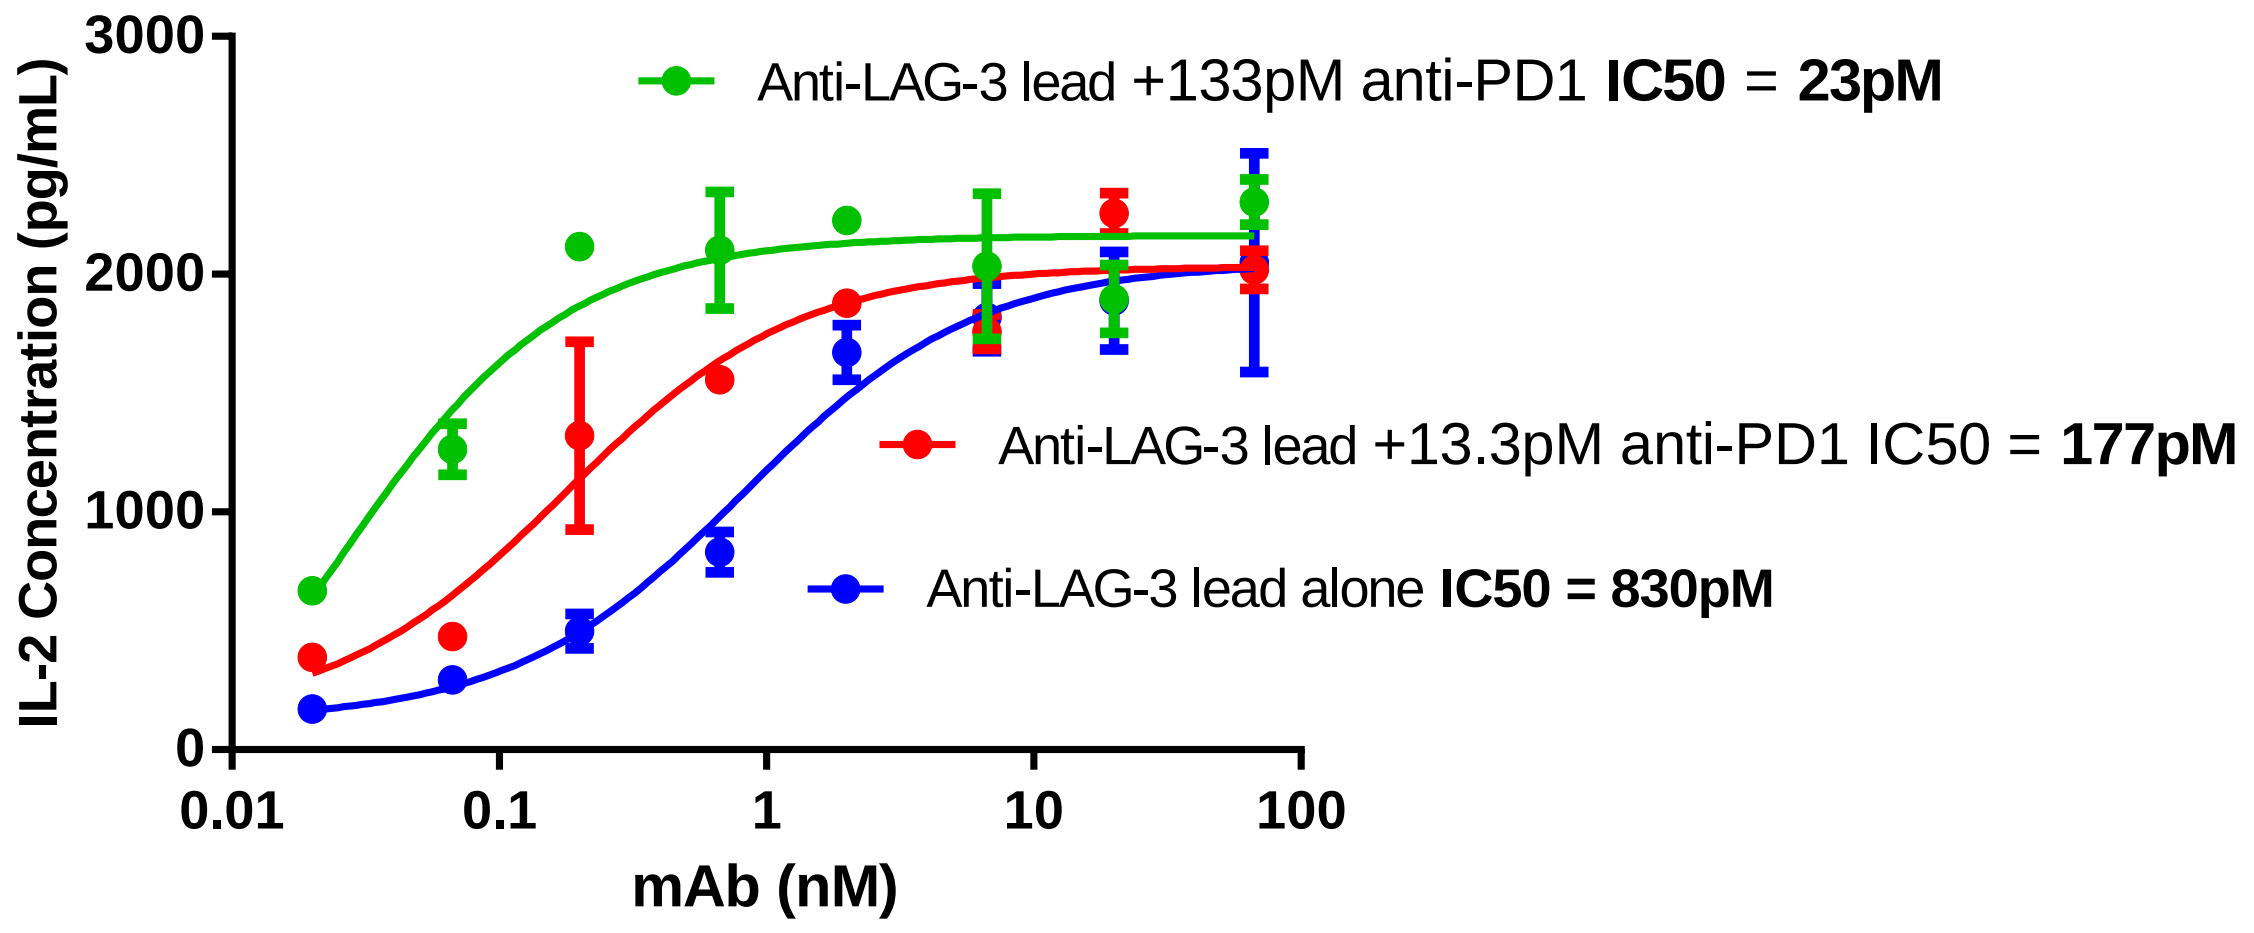
Anti-LAG-3 lead +133pM anti-PD1 IC50 = 23pM
Anti-LAG-3 lead +13.3pM anti-PD1 IC50 = 177pM
Anti-LAG-3 lead alone IC50 = 830pM
IL-2 Concentration (pg/mL)
mAb (nM)

Isolated peripheral blood monocytes from a human donor were differentiated into dendritic cells and then mixed with CD4+ T-cells isolated from a second donor. IL-2 levels were measured after 48 hours. The anti-LAG-3 mAb was combined with an anti-PD-1 mAb. PD-1 was able to decrease the anti-LAG-3 EC50.

Conclusions

- **A high affinity anti-human LAG-3 antibody has been generated by humanization of a mouse monoclonal Ab coupled with *in vitro* somatic hypermutation**
- **LAG-3 inhibition with an anti-LAG-3 antibody displays potent activity alone and in combination with an anti-PD-1 antibody in an MLR assay**
- **Surrogate anti-PD-1 and anti-LAG-3 antibodies have activity in the MC38 and Colon26 tumor models alone and increased anti-tumor activity in combination**
- **These data suggests co-blockade of PD-1 and LAG-3 is worthy of further investigation**